THE CFPC SECTION of RESEARCHERS

Preparation for Research Education/Excitement/Enhancement/Engagement in Practice (PRE^{E}P) Action Group:

Making Research A Core Component of Family Medicine Training, Scholarship, and Clinical Practice

Authors: Vivian Ramsden, RN, PhD, MCFP (Hon); Alan Katz, MBChB, CCFP, MSc, FCFP;
Cheryl Levitt, MBBCh, CCFP, FCFP

THE COLLEGE OF FAMILY PHYSICIANS OF CANADA
LE COLLÈGE DES MÉDECINS DE FAMILLE DU CANADA

The Section of Researchers' vision, as described in its Blueprint for Family Medicine Research Success 2012-2017 and in the College of Family Physicians of Canada's Strategic Plan 2013-2017, is to ensure that research is a core component of family medicine training, scholarship, and clinical practice. The Preparation for Research Education/Excitement/Enhancement/Engagement in Practice (PRE^{E}P) Action Group has been established to help build the research education, capacity, and capability of the family medicine community. The PRE^{E}P Action Group Planning Committee, which consisted of about 20 members, associate members and CFPC leaders, developed a vision and practical strategies for implementation.

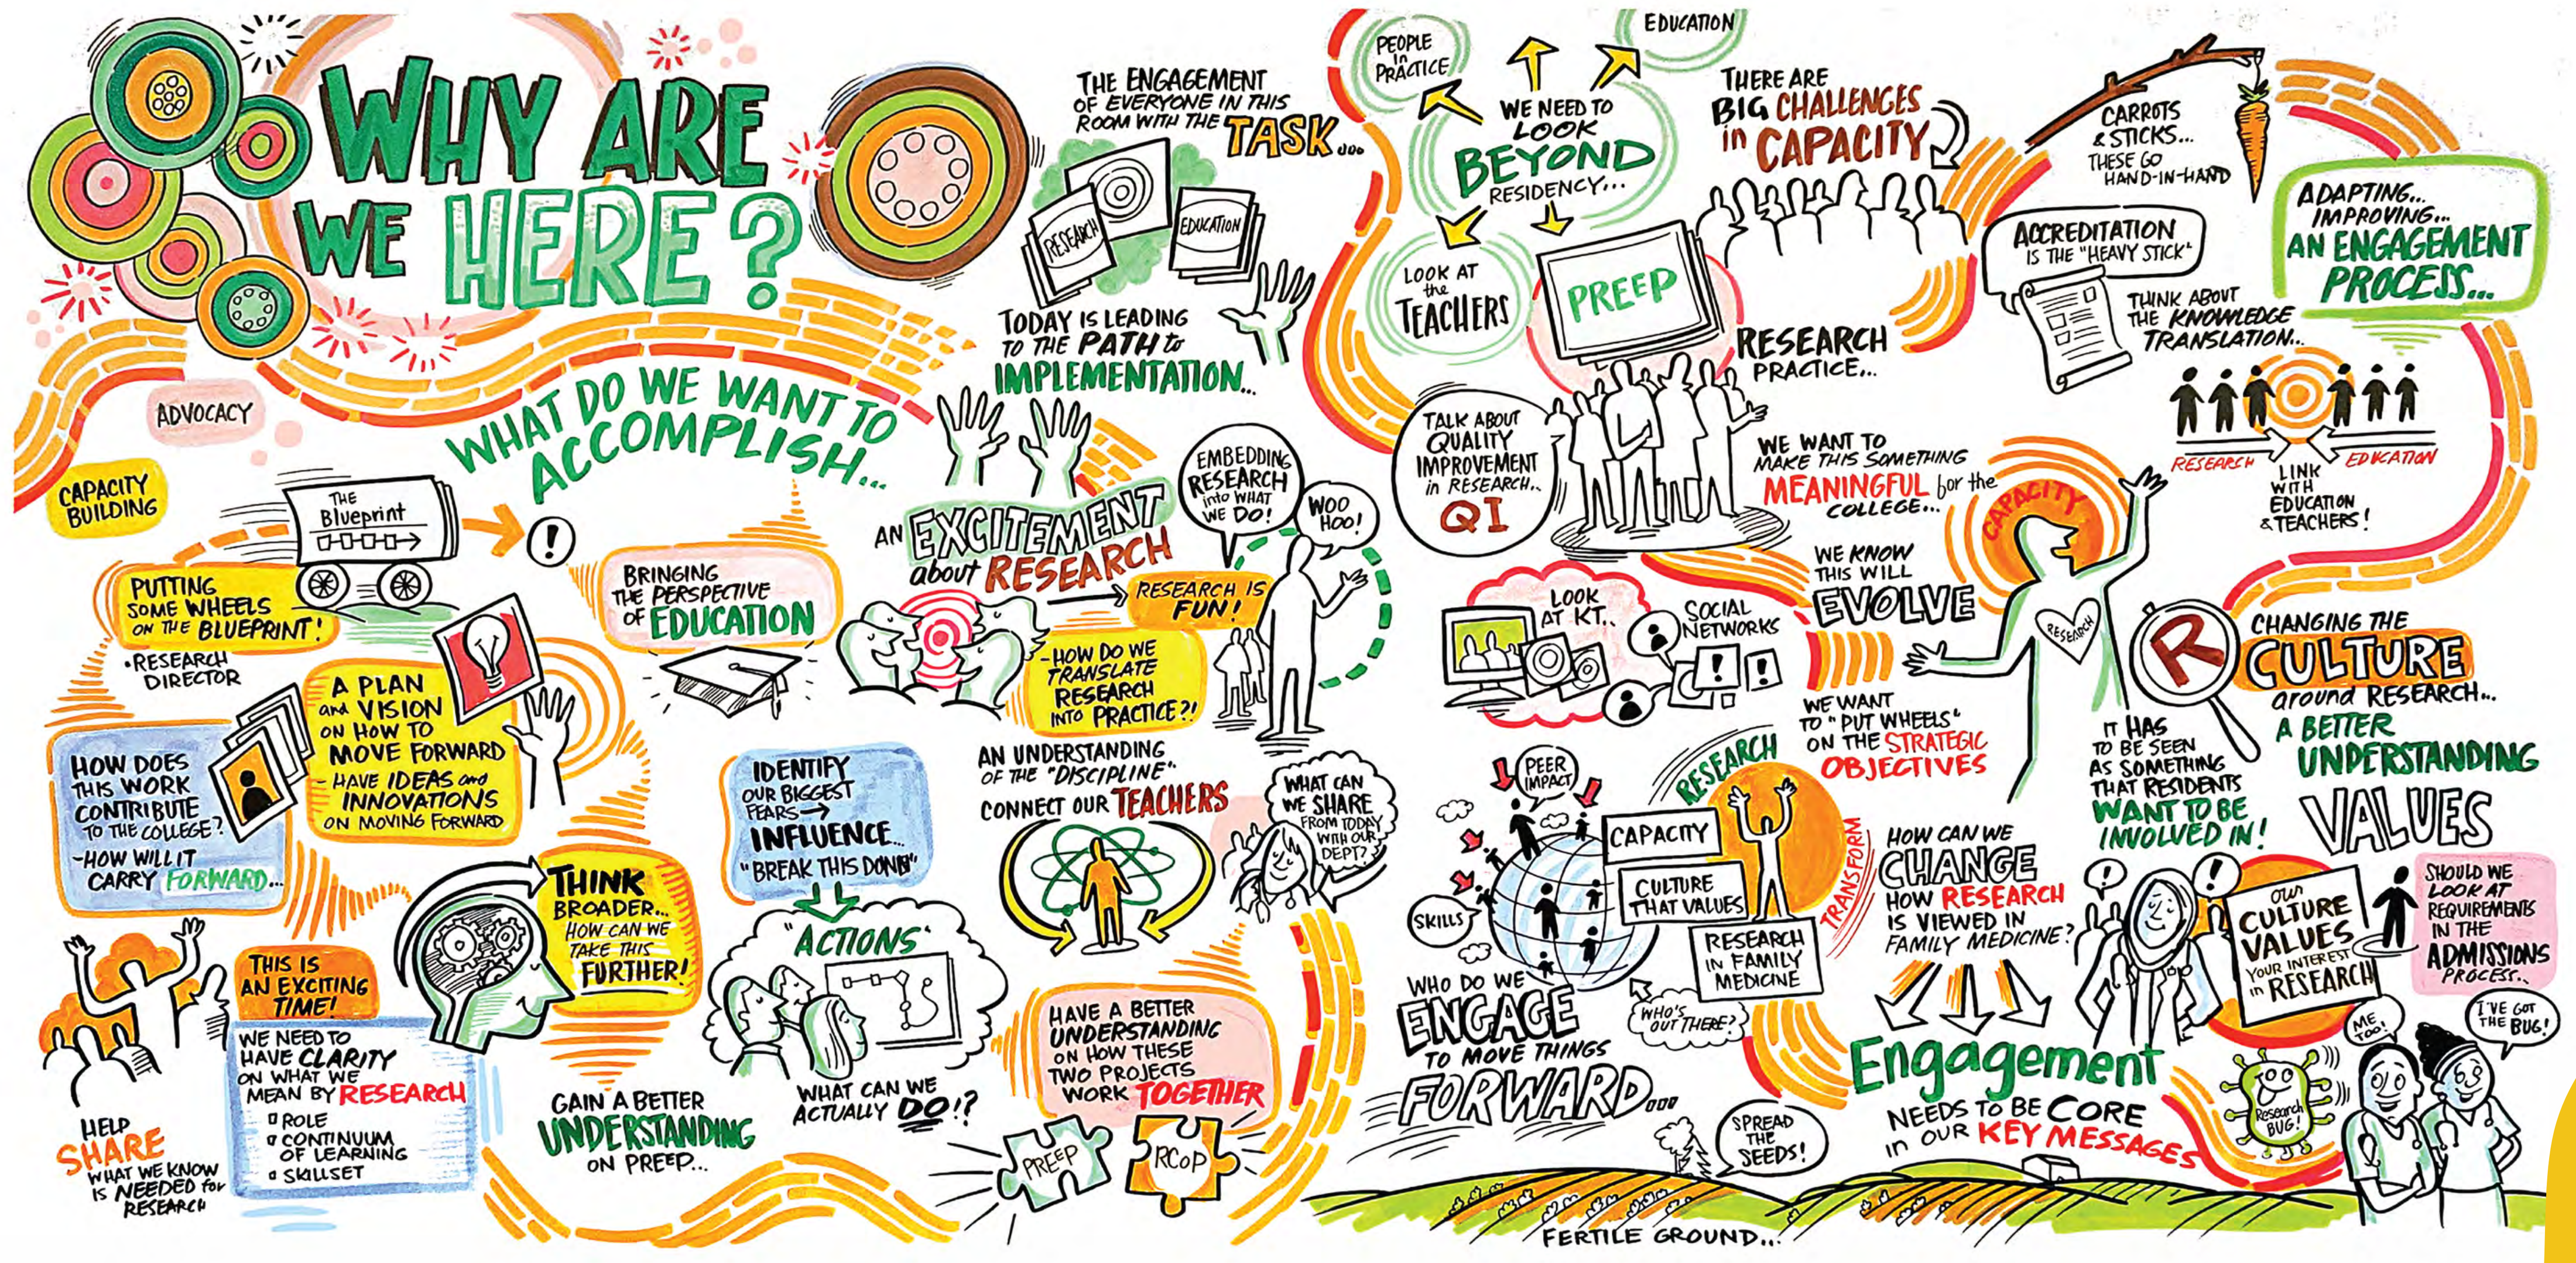
WHY ARE WE HERE?
WHAT DO WE WANT TO ACCOMPLISH...
ADVOCACY
CAPACITY BUILDING
PUTTING SOME WHEELS ON THE BLUEPRINT!
• RESEARCH DIRECTOR
A PLAN and VISION ON HOW TO MOVE FORWARD
HAVE IDEAS and INNOVATIONS ON MOVING FORWARD
HOW DOES THIS WORK CONTRIBUTE TO THE COLLEGE?
-HOW WILL IT CARRY FORWARD...
THIS IS AN EXCITING TIME!
WE NEED TO HAVE CLARITY ON WHAT WE MEAN BY RESEARCH
ROLE
CONTINUUM OF LEARNING
SKILLSET
HELP SHARE WHAT WE KNOW IS NEEDED for RESEARCH
THINK BROADER... HOW CAN WE TAKE THIS FURTHER!
GAIN A BETTER UNDERSTANDING ON PREEP...
BRINGING THE PERSPECTIVE OF EDUCATION
IDENTIFY OUR BIGGEST FEARS → INFLUENCE... "BREAK THIS DOWN"
"ACTIONS"
WHAT CAN WE ACTUALLY DO!?
TODAY IS LEADING TO THE PATH to IMPLEMENTATION...
AN EXCITEMENT about RESEARCH
RESEARCH IS FUN!
HOW DO WE TRANSLATE RESEARCH INTO PRACTICE?!
EMBEDDING RESEARCH into WHAT WE DO!
WOO HOO!
AN UNDERSTANDING OF THE "DISCIPLINE". CONNECT OUR TEACHERS
WHAT CAN WE SHARE FROM TODAY WITH OUR DEPT?
HAVE A BETTER UNDERSTANDING ON HOW THESE TWO PROJECTS WORK TOGETHER
PREEP
RCoP
THE ENGAGEMENT OF EVERYONE IN THIS ROOM WITH THE TASK...
PEOPLE in PRACTICE
EDUCATION
WE NEED TO LOOK BEYOND RESIDENCY...
LOOK AT the TEACHERS
PREEP
RESEARCH PRACTICE...
TALK ABOUT QUALITY IMPROVEMENT in RESEARCH... QI
THERE ARE BIG CHALLENGES in CAPACITY
WE WANT TO MAKE THIS SOMETHING MEANINGFUL for the COLLEGE...
CARROTS & STICKS... THESE GO HAND-IN-HAND
ACCREDITATION IS THE "HEAVY STICK"
THINK ABOUT THE KNOWLEDGE TRANSLATION...
ADAPTING... IMPROVING... AN ENGAGEMENT PROCESS...
RESEARCH
EDUCATION
LINK WITH EDUCATION & TEACHERS!
LOOK AT KT...
SOCIAL NETWORKS
WE KNOW THIS WILL EVOLVE
CAPACITY
WE WANT TO "PUT WHEELS" ON THE STRATEGIC OBJECTIVES
PEER IMPACT
SKILLS
CAPACITY
CULTURE THAT VALUES
RESEARCH IN FAMILY MEDICINE
TRANSFORM
WHO DO WE ENGAGE TO MOVE THINGS FORWARD...
WHO'S OUT THERE?
SPREAD THE SEEDS!
HOW CAN WE CHANGE HOW RESEARCH IS VIEWED IN FAMILY MEDICINE?
IT HAS TO BE SEEN AS SOMETHING THAT RESIDENTS WANT TO BE INVOLVED IN!
CHANGING THE CULTURE around RESEARCH...
A BETTER UNDERSTANDING
VALUES
OUR CULTURE VALUES YOUR INTEREST in RESEARCH
SHOULD WE LOOK AT REQUIREMENTS IN THE ADMISSIONS PROCESS...
ME TOO!
I'VE GOT THE BUG!
Research BUG!
Engagement NEEDS TO BE CORE in OUR KEY MESSAGES
FERTILE GROUND...

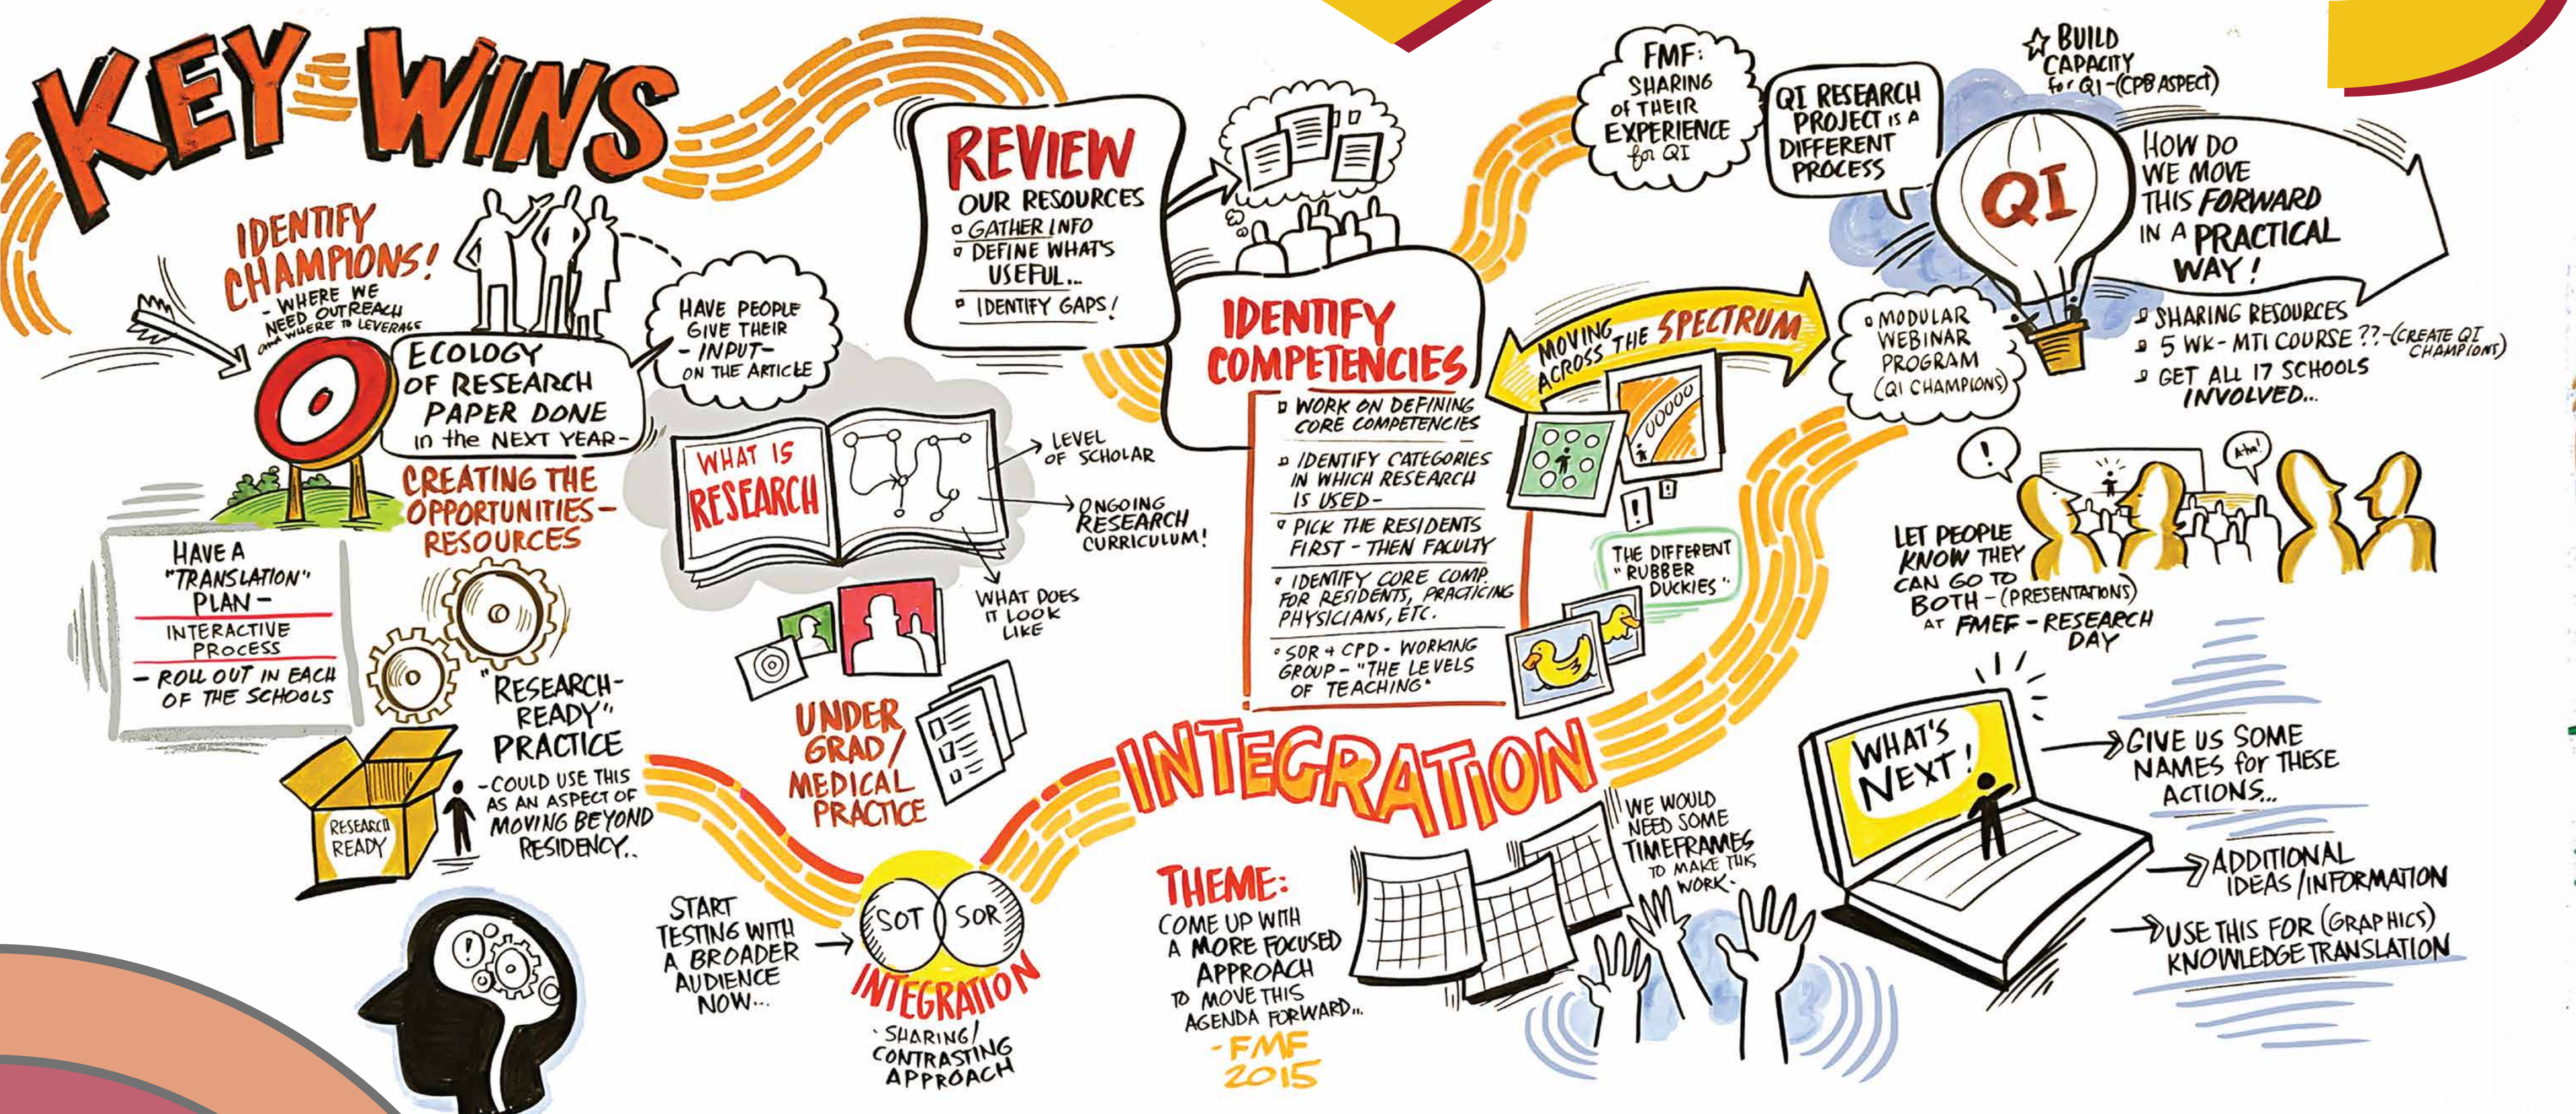
KEY WINS
IDENTIFY CHAMPIONS! - WHERE WE NEED OUTREACH and WHERE TO LEVERAGE
ECOLOGY OF RESEARCH PAPER DONE in the NEXT YEAR-
HAVE PEOPLE GIVE THEIR - INPUT - ON THE ARTICLE
CREATING THE OPPORTUNITIES - RESOURCES
HAVE A "TRANSLATION" PLAN -
INTERACTIVE PROCESS
- ROLL OUT IN EACH OF THE SCHOOLS
"RESEARCH-READY" PRACTICE
- COULD USE THIS AS AN ASPECT OF MOVING BEYOND RESIDENCY...
RESEARCH READY
WHAT IS RESEARCH
LEVEL OF SCHOLAR
ONGOING RESEARCH CURRICULUM!
WHAT DOES IT LOOK LIKE
UNDER GRAD/ MEDICAL PRACTICE
START TESTING WITH A BROADER AUDIENCE NOW...
SOT SOR
INTEGRATION
SHARING/ CONTRASTING APPROACH
REVIEW OUR RESOURCES
GATHER INFO
DEFINE WHAT'S USEFUL...
IDENTIFY GAPS!
IDENTIFY COMPETENCIES
WORK ON DEFINING CORE COMPETENCIES
IDENTIFY CATEGORIES IN WHICH RESEARCH IS USED -
PICK THE RESIDENTS FIRST - THEN FACULTY
IDENTIFY CORE COMP FOR RESIDENTS, PRACTICING PHYSICIANS, ETC.
SOR + CPD - WORKING GROUP - "THE LEVELS OF TEACHING"
MOVING ACROSS THE SPECTRUM
THE DIFFERENT "RUBBER DUCKIES"
INTEGRATION
THEME: COME UP WITH A MORE FOCUSED APPROACH TO MOVE THIS AGENDA FORWARD... - FMF 2015
WE WOULD NEED SOME TIMEFRAMES TO MAKE THIS WORK
FMF: SHARING OF THEIR EXPERIENCE for QI
QI RESEARCH PROJECT IS A DIFFERENT PROCESS
BUILD CAPACITY for QI - (CPD ASPECT)
QI
HOW DO WE MOVE THIS FORWARD IN A PRACTICAL WAY!
MODULAR WEBINAR PROGRAM (QI CHAMPIONS)
SHARING RESOURCES
5 WK - MTI COURSE ?? - (CREATE QI CHAMPIONS)
GET ALL 17 SCHOOLS INVOLVED...
LET PEOPLE KNOW THEY CAN GO TO BOTH - (PRESENTATIONS) AT FMEF - RESEARCH DAY
Aha!
WHAT'S NEXT!
GIVE US SOME NAMES for THESE ACTIONS...
ADDITIONAL IDEAS / INFORMATION
USE THIS FOR (GRAPHICS) KNOWLEDGE TRANSLATION

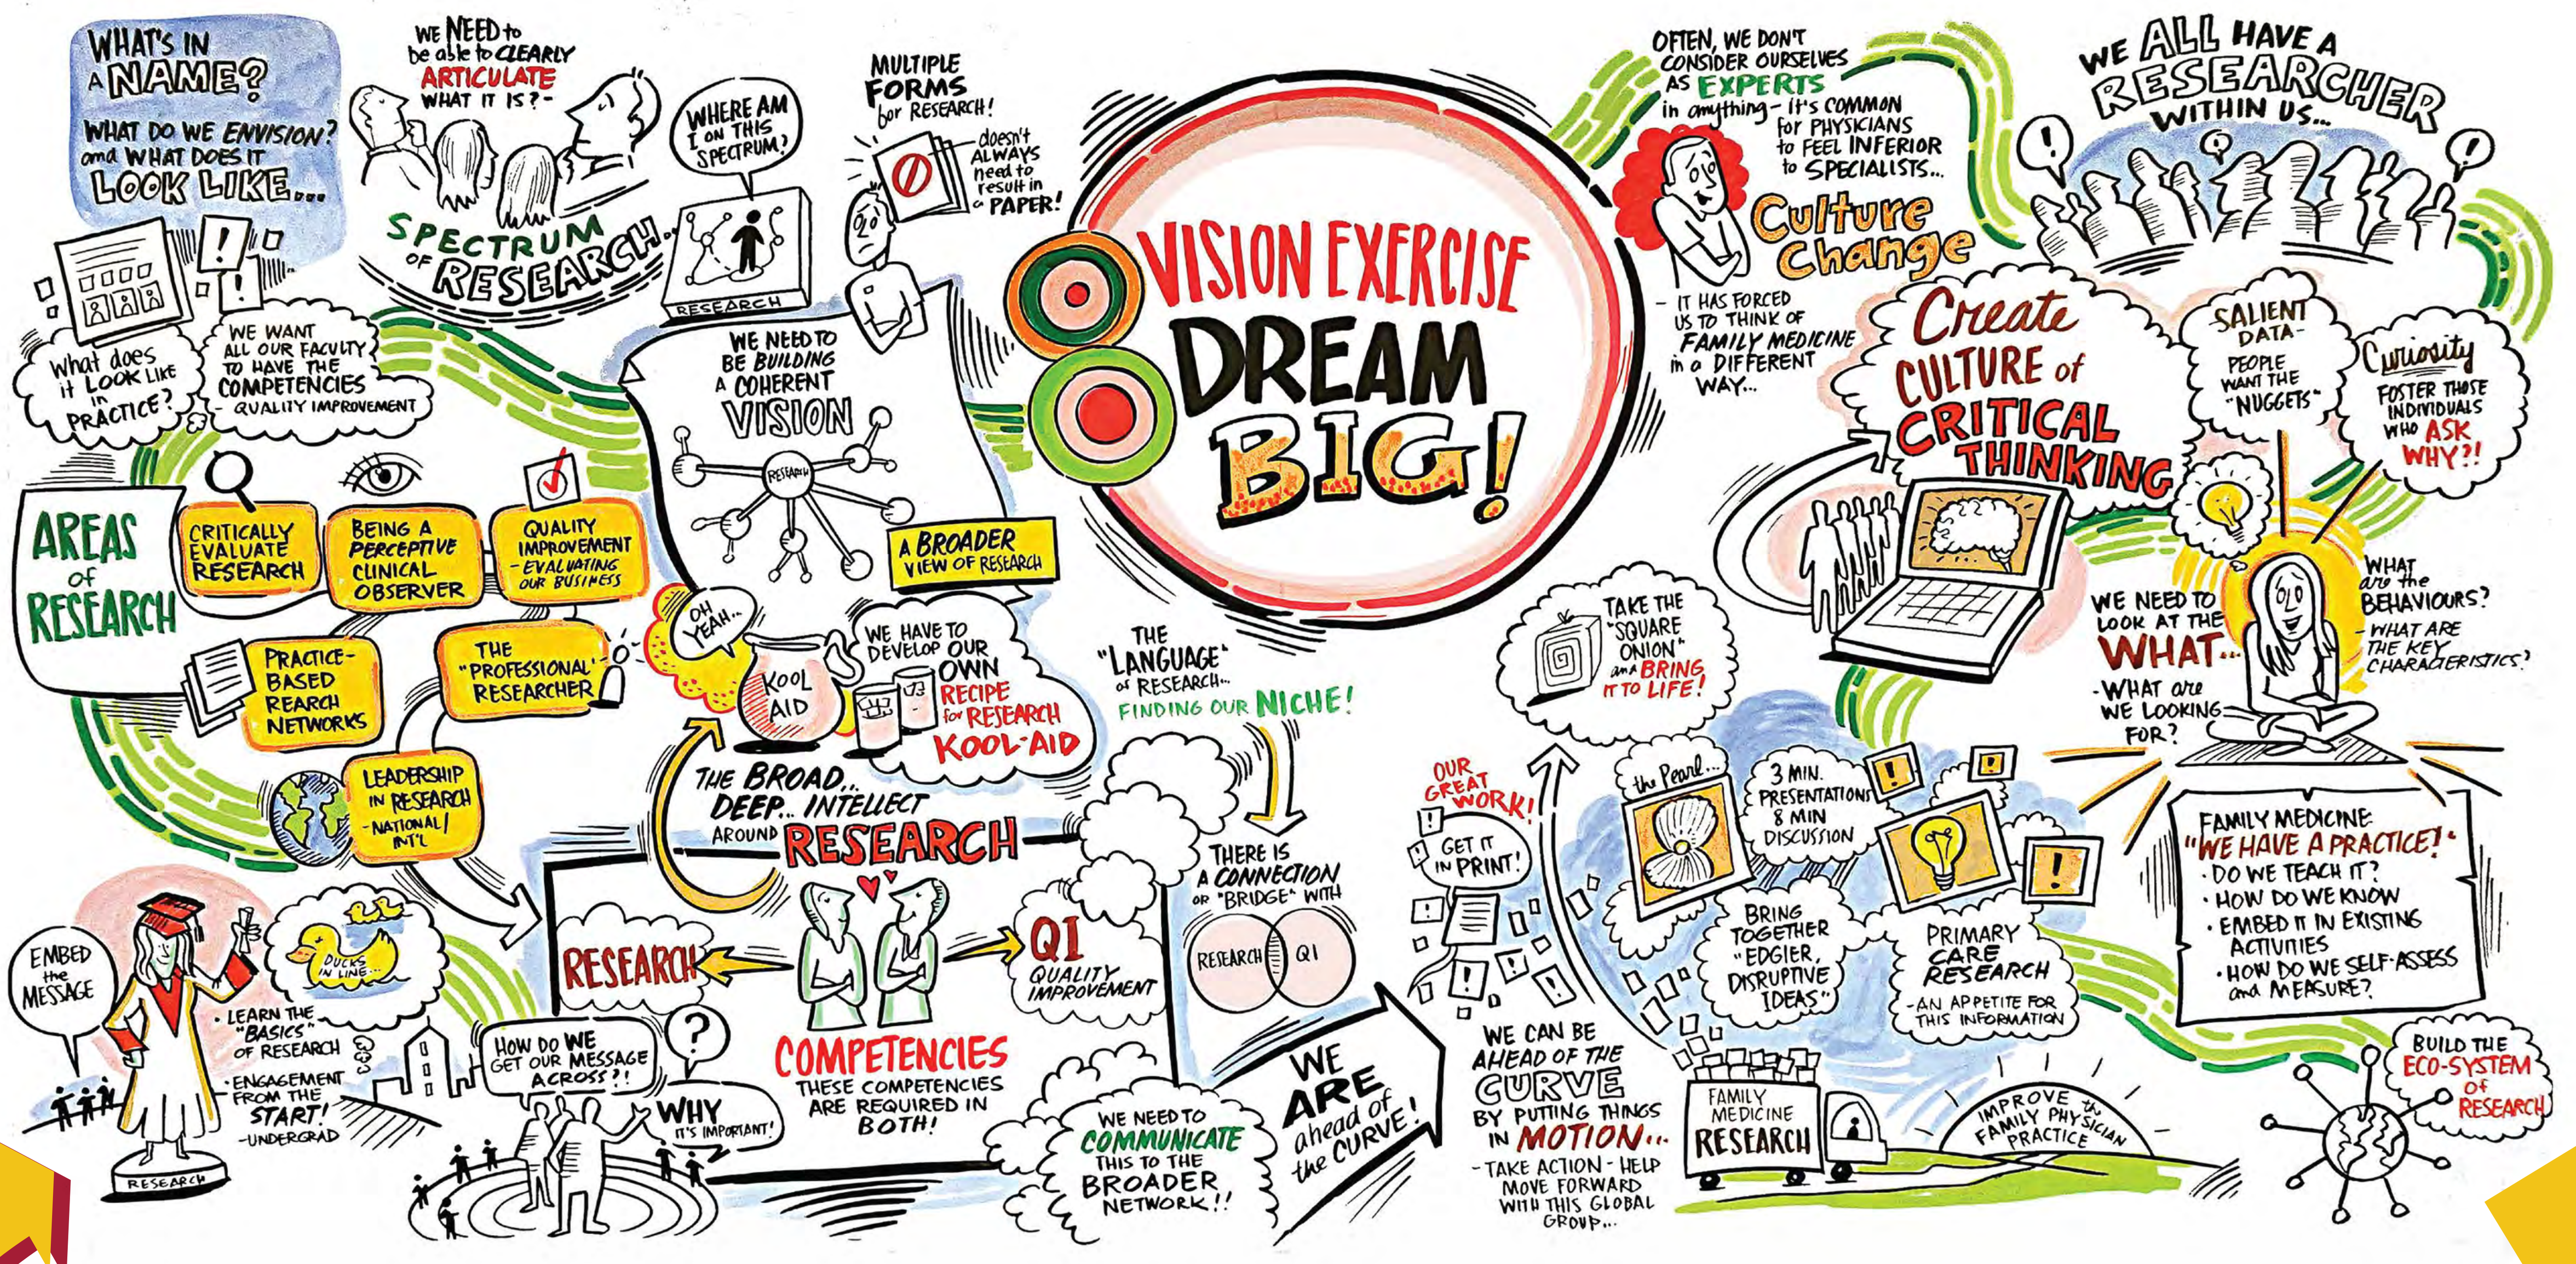
WHAT'S IN A NAME?
WHAT DO WE ENVISION? and WHAT DOES IT LOOK LIKE...
WE NEED to be able to CLEARLY ARTICULATE WHAT IT IS?
WHERE AM I ON THIS SPECTRUM?
SPECTRUM of RESEARCH
MULTIPLE FORMS for RESEARCH! - doesn't ALWAYS need to result in a PAPER!
What does it LOOK LIKE in PRACTICE?
WE WANT ALL OUR FACULTY TO HAVE THE COMPETENCIES - QUALITY IMPROVEMENT
WE NEED TO BE BUILDING A COHERENT VISION
AREAS of RESEARCH
CRITICALLY EVALUATE RESEARCH
BEING A PERCEPTIVE CLINICAL OBSERVER
QUALITY IMPROVEMENT - EVALUATING OUR BUSINESS
A BROADER VIEW OF RESEARCH
PRACTICE-BASED REARCH NETWORKS
THE "PROFESSIONAL" RESEARCHER
OH YEAH...
KOOL AID
WE HAVE TO DEVELOP OUR OWN RECIPE for RESEARCH KOOL-AID
LEADERSHIP IN RESEARCH - NATIONAL / INT'L
THE BROAD... DEEP... INTELLECT AROUND RESEARCH
EMBED the MESSAGE
LEARN THE "BASICS" OF RESEARCH
ENGAGEMENT FROM THE START! - UNDERGRAD
DUCKS IN LINE...
RESEARCH
HOW DO WE GET OUR MESSAGE ACROSS?!
WHY IT'S IMPORTANT!
COMPETENCIES THESE COMPETENCIES ARE REQUIRED IN BOTH!
QI QUALITY IMPROVEMENT
THE "LANGUAGE" of RESEARCH... FINDING OUR NICHE!
THERE IS A CONNECTION or "BRIDGE" WITH
RESEARCH QI
WE NEED TO COMMUNICATE THIS TO THE BROADER NETWORK!!
WE ARE ahead of the CURVE!
VISION EXERCISE DREAM BIG!
OFTEN, WE DON'T CONSIDER OURSELVES AS EXPERTS in anything - it's COMMON for PHYSICIANS to FEEL INFERIOR to SPECIALISTS...
Culture Change
- IT HAS FORCED US TO THINK OF FAMILY MEDICINE in a DIFFERENT WAY...
WE ALL HAVE A RESEARCHER WITHIN US...
Create CULTURE of CRITICAL THINKING
SALIENT DATA - PEOPLE WANT THE "NUGGETS"
Curiosity FOSTER THOSE INDIVIDUALS WHO ASK WHY?!
TAKE THE "SQUARE ONION" and BRING IT TO LIFE!
WE NEED TO LOOK AT THE WHAT...
- WHAT are WE LOOKING FOR?
WHAT are the BEHAVIOURS?
- WHAT ARE THE KEY CHARACTERISTICS?
OUR GREAT WORK!
GET IT IN PRINT!
the Pearl...
3 MIN. PRESENTATIONS 8 MIN DISCUSSION
BRING TOGETHER "EDGIER, DISRUPTIVE IDEAS"
PRIMARY CARE RESEARCH - AN APPETITE FOR THIS INFORMATION
FAMILY MEDICINE: "WE HAVE A PRACTICE!"
· DO WE TEACH IT?
· HOW DO WE KNOW
· EMBED IT IN EXISTING ACTIVITIES
· HOW DO WE SELF-ASSESS and MEASURE?
WE CAN BE AHEAD OF THE CURVE BY PUTTING THINGS IN MOTION...
- TAKE ACTION - HELP MOVE FORWARD WITH THIS GLOBAL GROUP...
FAMILY MEDICINE RESEARCH
IMPROVE the FAMILY PHYSICIAN PRACTICE
BUILD THE ECO-SYSTEM OF RESEARCH

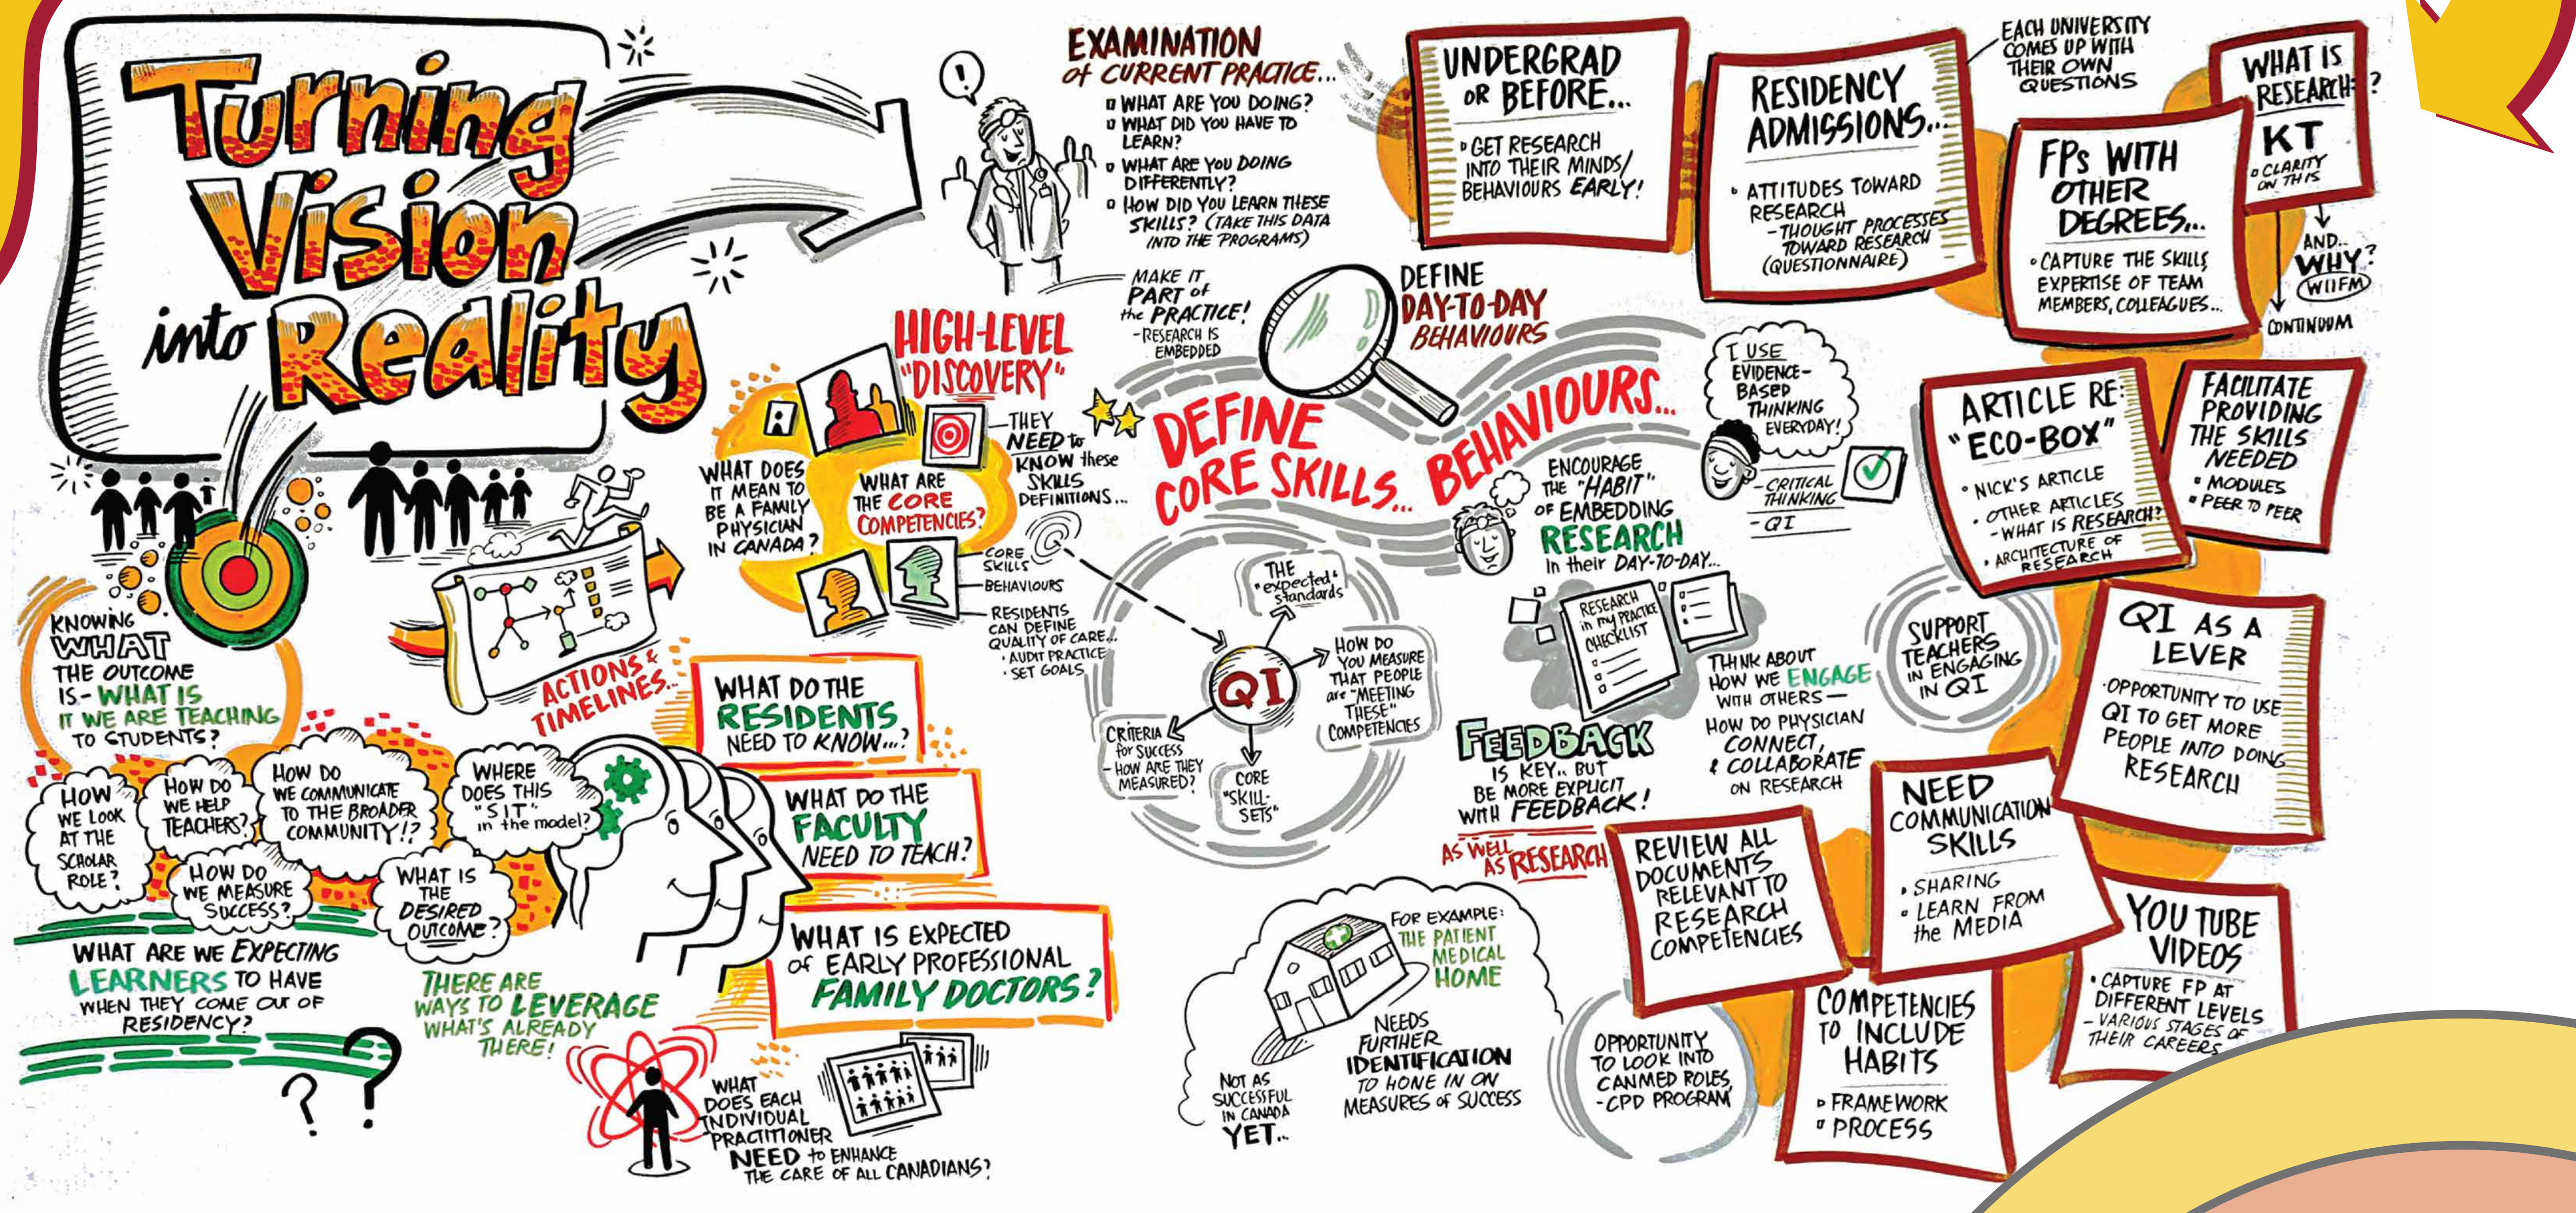
Turning Vision into Reality
KNOWING WHAT THE OUTCOME IS - WHAT IS IT WE ARE TEACHING TO STUDENTS?
HOW WE LOOK AT THE SCHOLAR ROLE?
HOW DO WE HELP TEACHERS?
HOW DO WE COMMUNICATE TO THE BROADER COMMUNITY!?
WHERE DOES THIS "SIT" in the model?
HOW DO WE MEASURE SUCCESS?
WHAT IS THE DESIRED OUTCOME?
WHAT ARE WE EXPECTING LEARNERS TO HAVE WHEN THEY COME OUT OF RESIDENCY?
THERE ARE WAYS TO LEVERAGE WHAT'S ALREADY THERE!
ACTIONS & TIMELINES...
WHAT DOES IT MEAN TO BE A FAMILY PHYSICIAN IN CANADA?
WHAT ARE THE CORE COMPETENCIES?
HIGH-LEVEL "DISCOVERY"
- THEY NEED to KNOW these SKILLS DEFINITIONS...
CORE SKILLS
BEHAVIOURS
RESIDENTS CAN DEFINE QUALITY OF CARE... · AUDIT PRACTICE · SET GOALS
WHAT DO THE RESIDENTS NEED TO KNOW...?
WHAT DO THE FACULTY NEED TO TEACH?
WHAT IS EXPECTED OF EARLY PROFESSIONAL FAMILY DOCTORS?
WHAT DOES EACH INDIVIDUAL PRACTITIONER NEED to ENHANCE THE CARE OF ALL CANADIANS?
EXAMINATION of CURRENT PRACTICE...
WHAT ARE YOU DOING?
WHAT DID YOU HAVE TO LEARN?
WHAT ARE YOU DOING DIFFERENTLY?
HOW DID YOU LEARN THESE SKILLS? (TAKE THIS DATA INTO THE PROGRAMS)
MAKE IT PART of the PRACTICE! - RESEARCH IS EMBEDDED
DEFINE DAY-TO-DAY BEHAVIOURS
DEFINE CORE SKILLS... BEHAVIOURS...
THE "expected" standards
HOW DO YOU MEASURE THAT PEOPLE are "MEETING THESE" COMPETENCIES
QI
CRITERIA for SUCCESS - HOW ARE THEY MEASURED?
CORE "SKILL-SETS"
ENCOURAGE THE "HABIT" of EMBEDDING RESEARCH In their DAY-TO-DAY...
RESEARCH in my PRACTICE CHECKLIST
I USE EVIDENCE-BASED THINKING EVERYDAY!
- CRITICAL THINKING
- QI
FEEDBACK IS KEY... BUT BE MORE EXPLICIT WITH FEEDBACK!
AS WELL AS RESEARCH
THINK ABOUT HOW WE ENGAGE WITH OTHERS - HOW DO PHYSICIAN CONNECT, & COLLABORATE ON RESEARCH
SUPPORT TEACHERS IN ENGAGING IN QI
FOR EXAMPLE: THE PATIENT MEDICAL HOME
NEEDS FURTHER IDENTIFICATION TO HONE IN ON MEASURES of SUCCESS
NOT AS SUCCESSFUL IN CANADA YET...
OPPORTUNITY TO LOOK INTO CANMED ROLES, -CPD PROGRAM
UNDERGRAD OR BEFORE...
· GET RESEARCH INTO THEIR MINDS/ BEHAVIOURS EARLY!
RESIDENCY ADMISSIONS...
· ATTITUDES TOWARD RESEARCH - THOUGHT PROCESSES TOWARD RESEARCH (QUESTIONNAIRE)
EACH UNIVERSITY COMES UP WITH THEIR OWN QUESTIONS
FPs WITH OTHER DEGREES...
· CAPTURE THE SKILLS EXPERTISE OF TEAM MEMBERS, COLLEAGUES...
WHAT IS RESEARCH?
KT
· CLARITY ON THIS
AND... WHY? (WIIFM)
CONTINUUM
ARTICLE RE: "ECO-BOX"
· NICK'S ARTICLE
· OTHER ARTICLES - WHAT IS RESEARCH?
· ARCHITECTURE OF RESEARCH
FACILITATE PROVIDING THE SKILLS NEEDED
· MODULES
· PEER TO PEER
QI AS A LEVER
· OPPORTUNITY TO USE QI TO GET MORE PEOPLE INTO DOING RESEARCH
REVIEW ALL DOCUMENTS RELEVANT TO RESEARCH COMPETENCIES
NEED COMMUNICATION SKILLS
· SHARING
· LEARN FROM the MEDIA
COMPETENCIES TO INCLUDE HABITS
· FRAMEWORK
· PROCESS
YOU TUBE VIDEOS
· CAPTURE FP AT DIFFERENT LEVELS
- VARIOUS STAGES OF THEIR CAREERS

CFPC Section of Researchers' Blueprint Objective 2:
Advocate for and support an increase in research capacity and capability in academic departments and teaching sites.

CFPC Strategic Plan Goal 4:
Advance the discipline of family medicine through innovation, research, and transfer of knowledge.